Poster# 1

AMCP Nexus
October 26-29, 2026
San Diego, CA, USA

Cost-Effectiveness of Symbravo® (MoSEIC™ Meloxicam and Rizatriptan) Compared With Gepants for the Acute Treatment of Migraine in Adults in the United States

Jessica Ailani,[1,2] Christopher N. Graham,[3] Amanda Erbe,[3] Todd Grinnell,[4] Ronnie DePue,[4] Yang Zhao[4]

[1]MedStar Georgetown University Hospital, McLean, VA, USA; [2]MedStar Georgetown Headache Center, McLean, VA, USA; [3]RTI Health Solutions, Durham, NC, USA; [4]Axsome Therapeutics, New York, NY, USA

Objective

To assess the cost-effectiveness of treatment with Symbravo® (MoSEIC™ meloxicam and rizatriptan [MxRz; previously AXS-07]) for patients with acute migraine compared with rimegepant, ubrogepant, and zavegepant from a US payer perspective

Introduction

- Migraine is a disabling neurologic condition that requires acute treatment. Existing treatments are often suboptimal in providing rapid and sustained relief of pain and associated symptoms, which can increase healthcare utilization and costs[1-8]
- In the post-triptan era, acute treatments for migraine include oral and nasal spray calcitonin gene-related peptide (CGRP) inhibitors (gepants; eg, rimegepant, ubrogepant, and zavegepant) and MxRz—a novel, multimechanistic oral medication comprising MoSEIC™ meloxicam and rizatriptan that is approved in the United States (US) for the treatment of migraine with or without aura in adults[9-11]
- Relative cost-effectiveness comparisons are helpful to estimate the economic value of available therapies, particularly given the increases in acute migraine treatment costs[21]
 - A network meta-analysis (NMA) found MxRz to be more efficacious than gepants[10,12-20]

Methods

Model Structure

- A semi-Markov model was developed following the Institute for Clinical and Economic Review (ICER) Acute Treatments for Migraine 2020 Report[22] with 4 health states to capture the costs and utilities associated with the treatment of acute migraine over 2-year (base case) and 1-year (scenario analysis) periods
 - The patient population reflected those enrolled in the phase 3 MOMENTUM clinical trial for MxRz (NCT03896009),[10] as these patients experienced moderate to severe migraine, similarly to patients in the gepant trials[10,12-20]

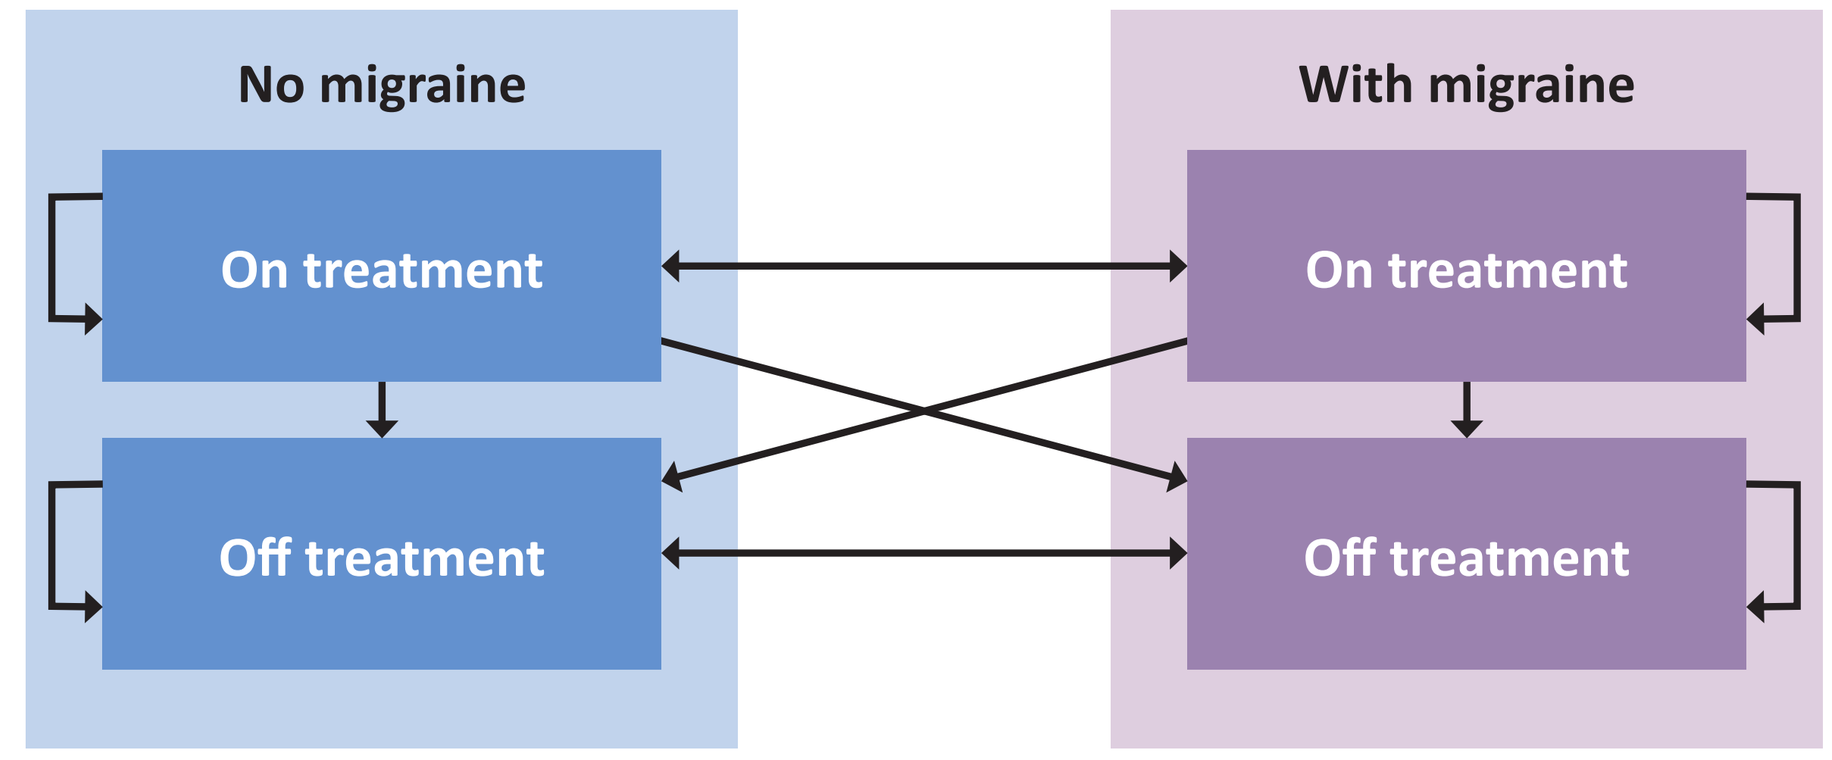
No migraine
On treatment
Off treatment
With migraine
On treatment
Off treatment

Model Inputs

Parameter/input	Base case/assumption
Cycle length	48 hours
Discount rate – costs and outcomes	3.0%
Cost year	2026 USD[a]
Drug use and costs	• Wholesale acquisition costs[b] • Assumed 50/50 distribution of 50-mg and 100-mg ubrogepant doses; second treatment dose accounted for only ubrogepant (20% of patients[15])
Treatment efficacy	• Efficacy endpoints included pain relief, pain freedom, absence of most bothersome symptoms, ability to function, and avoidance of rescue medication from 2 to 24 hours • Derived from Bayesian NMA of phase 3 study data for MxRz and gepants[10,12-20] • Cost and utility data extracted from the literature
Treatment discontinuation and adverse events	• Discontinuation rates due to lack of effect were assumed the same across all treatments (21.8%) and based on findings from a 12-month OLE study of lasmiditan, summarized by the ICER[22] • Discontinuation rates due to adverse events were 1.8% for MxRz,[23] 2.7% for rimegepant,[22] 2.2% (50 mg) and 2.7% (100 mg) for ubrogepant,[22] and 6.8% for zavegepant[24] • Adverse events based on PI[c]; last for 8 hours[22]
Healthcare resource utilization	• Physician visit, emergency department visit, and hospitalization costs based on utilization data[25] and efficacy results from Bayesian NMA[10,12-20] • Estimates based on the probability of resource use and the percentage of patients remaining in moderate or severe migraine pain at 12 hours for each treatment

ICER, Institute for Clinical and Economic Review; OLE, open-label extension; PI, prescribing information; USD, US dollar
[a]Costs adjusted to 2026 USD using the Personal Consumption Expenditures Index[26]
[b]Sourced from the Red Book[27]
[c]MxRz[11]; rimegepant[28]; ubrogepant[29]; zavegepant[30]

Outcomes

- Total costs and totals of the following outcomes were estimated: quality-adjusted life-years (QALYs), total hours of pain, pain freedom at 2 hours, sustained pain freedom from 2 to 24 hours, and avoidance of rescue medications from 2 to 24 hours
- Incremental cost-effectiveness ratios (ICERs) were calculated for the incremental cost per QALY gained and the incremental cost per hour of pain avoided
- Cost per outcomes was calculated for each comparator for the following outcomes: cost per responder for pain freedom at 2 hours and sustained pain freedom from 2 to 24 hours, cost per rescue medication avoided from 2 to 24 hours
- One-way sensitivity analyses (OWSAs) and probabilistic sensitivity analyses (PSAs) were conducted
 - OWSAs varied each model input either by using published or NMA confidence interval or by applying a ±20% range around the base-case values
 - PSAs assessed the impact of simultaneous variation in key parameters, using 1000 iterations where the model parameters were sampled from predefined probability distributions and a willingness-to-pay threshold of $100,000 per QALY gained, representing the maximum amount a payer is willing to spend for 1 additional QALY

Results

Total Costs, QALYs, and ICERs

Total Costs, 2-Year Time Horizon

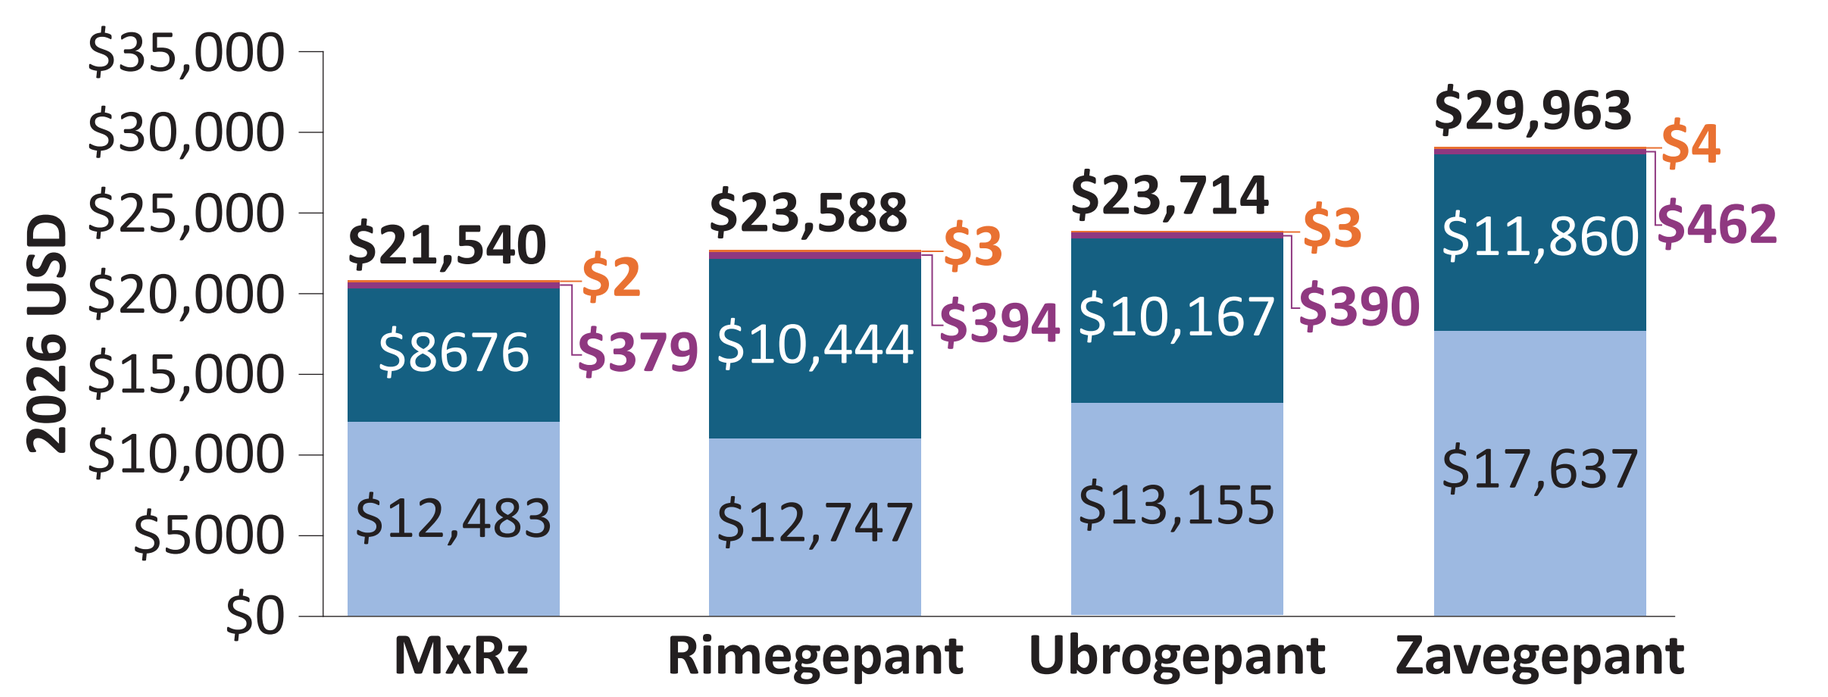
2026 USD
$35,000
$30,000
$25,000
$20,000
$15,000
$10,000
$5000
$0
$21,540 $2 $379 $8676 $12,483
$23,588 $3 $394 $10,444 $12,747
$23,714 $3 $390 $10,167 $13,155
$29,963 $4 $462 $11,860 $17,637
MxRz
Rimegepant
Ubrogepant
Zavegepant

Total Costs, 1-Year Time Horizon

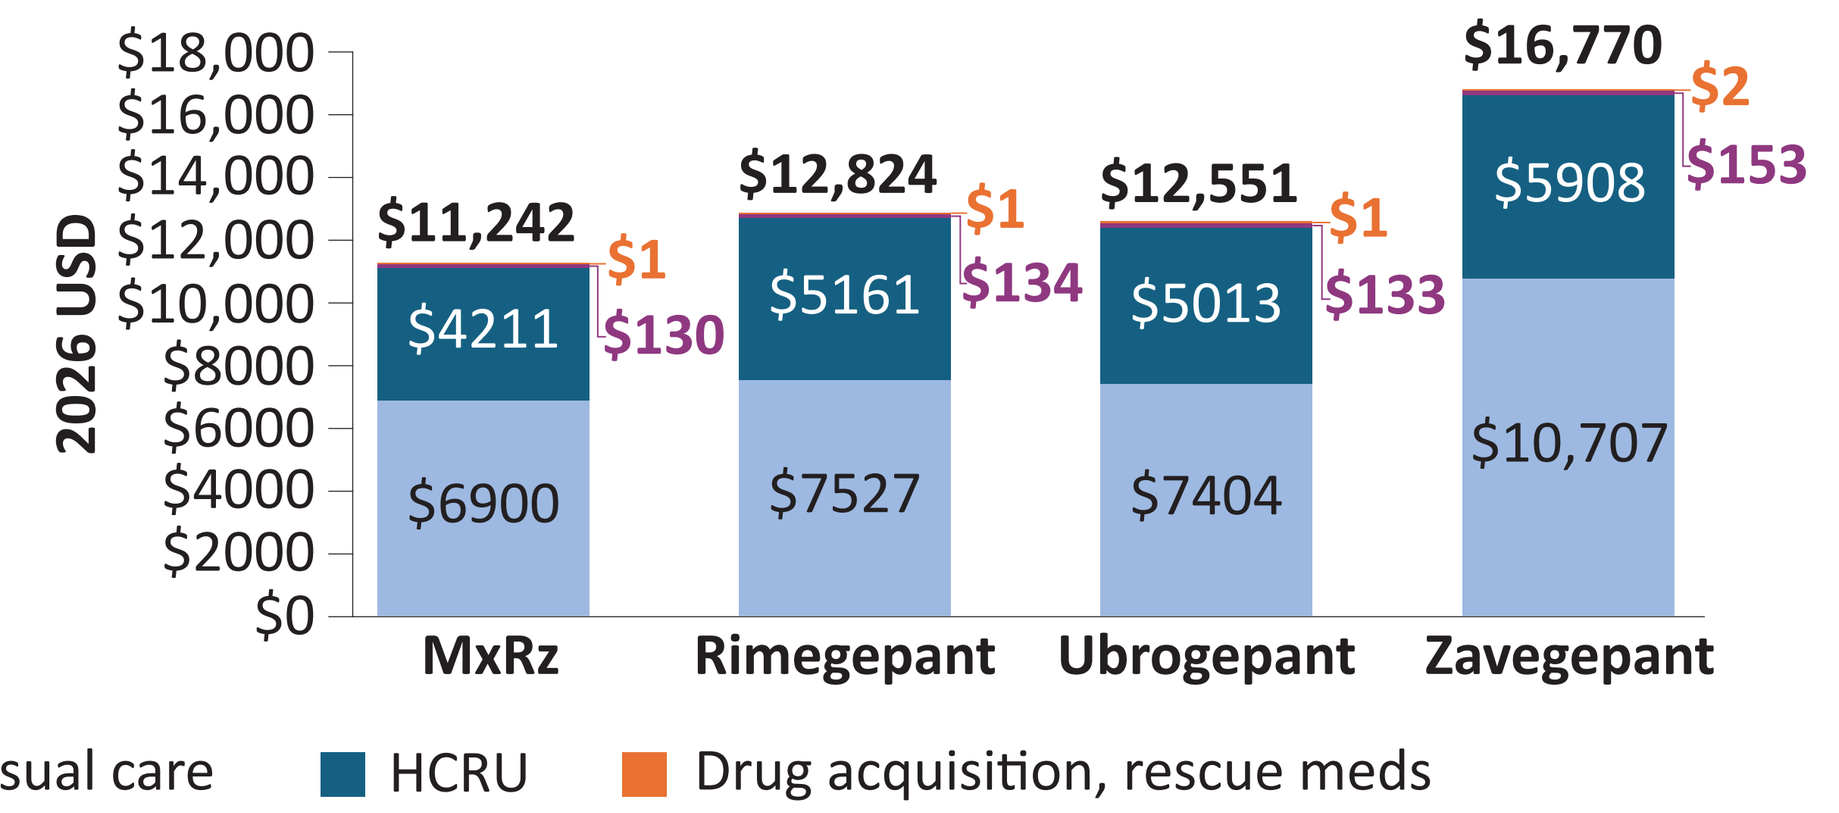
2026 USD
$18,000
$16,000
$14,000
$12,000
$10,000
$8000
$6000
$4000
$2000
$0
$11,242 $1 $130 $4211 $6900
$12,824 $1 $134 $5161 $7527
$12,551 $1 $133 $5013 $7404
$16,770 $2 $153 $5908 $10,707
MxRz
Rimegepant
Ubrogepant
Zavegepant
Drug acquisition, treatment
Drug acquisition, usual care
HCRU
Drug acquisition, rescue meds

- Treatment with MxRz over 2 years was associated with the lowest total cost ($21,540) compared with rimegepant ($23,588), ubrogepant ($23,714), and zavegepant ($29,963)
- As derived from the Bayesian NMA model[20]:
 - A significantly greater proportion of patients treated with MxRz were estimated to achieve pain freedom at 2 hours (34.3%) compared with gepants (approximately 20.0-21.0%)
 - MxRz was also estimated to demonstrate superior sustained pain freedom from 2 to 24 hours (24.0%) relative to rimegepant (16.0%), ubrogepant (13.3%), and zavegepant (12.3%)
- MxRz's fast onset of relief and lasting durability translated to an estimated fewer hours in pain, lower HCRU, and lower reliance on rescue medication (15.5% versus 18.0-27.9%), resulting in the lowest costs
- Similar findings were observed for the 1-year time horizon

Notes: Rescue medication cost assumed to be weighted average of the prior medications taken by ≥ 5% of participants in the MxRz group of the MOMENTUM trial, with over-the-counter medications (eg, ibuprofen, naproxen sodium, naproxen, acetylsalicylic acid, caffeine, paracetamol, and butalbital) incurred by the patient (ie, $0 to the health plan)

Totals may not sum exactly due to rounding

Estimated Total QALYs

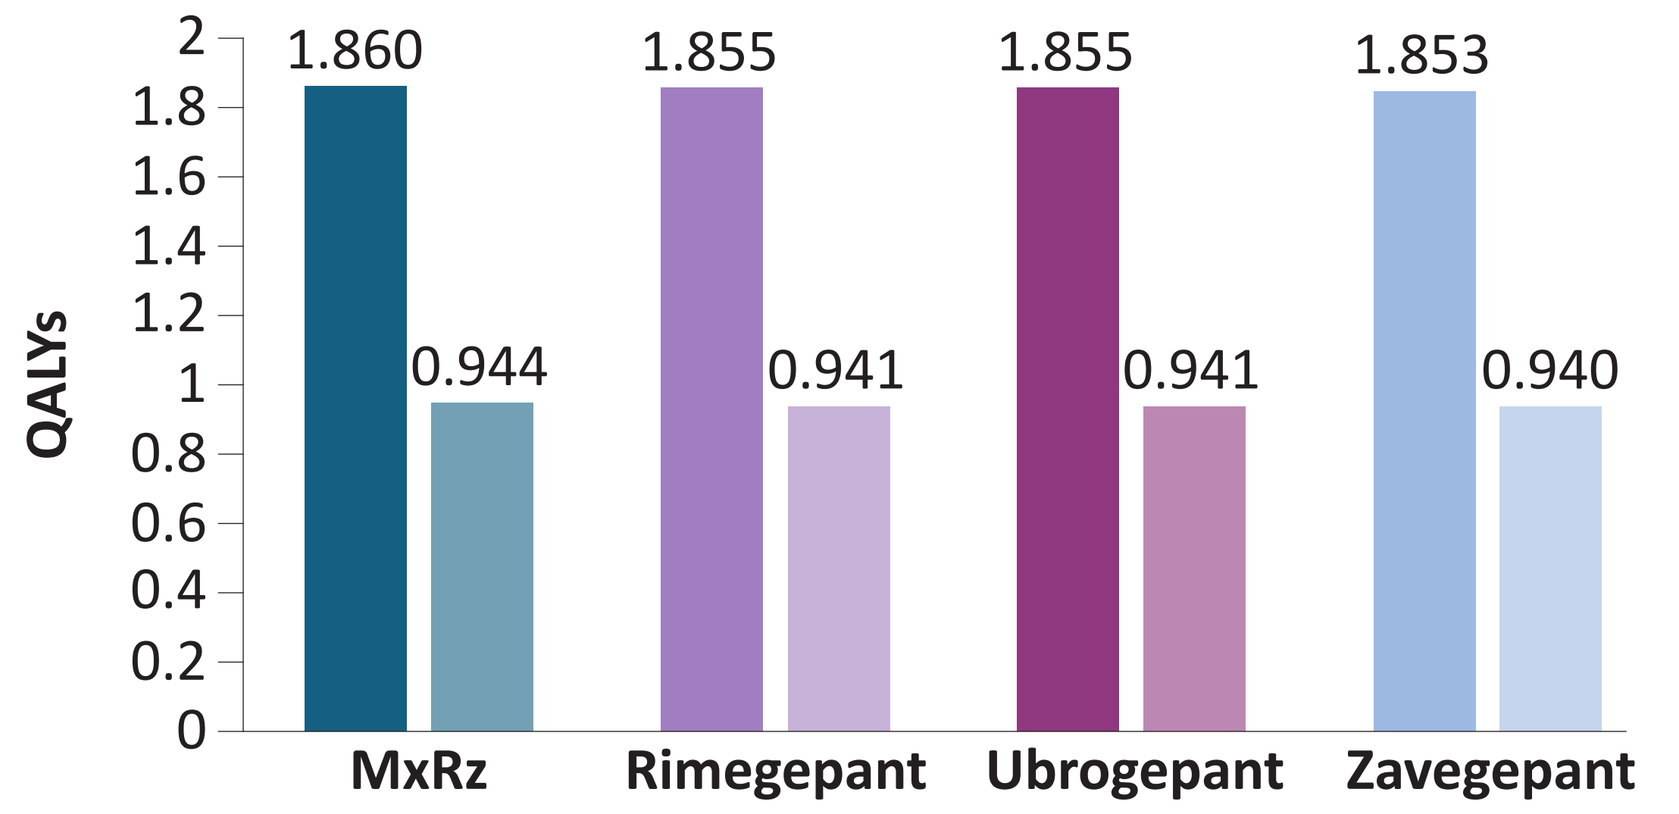
QALYs
2
1.8
1.6
1.4
1.2
1
0.8
0.6
0.4
0.2
0
1.860 0.944
1.855 0.941
1.855 0.941
1.853 0.940
MxRz
Rimegepant
Ubrogepant
Zavegepant

Estimated Total Hours of Pain

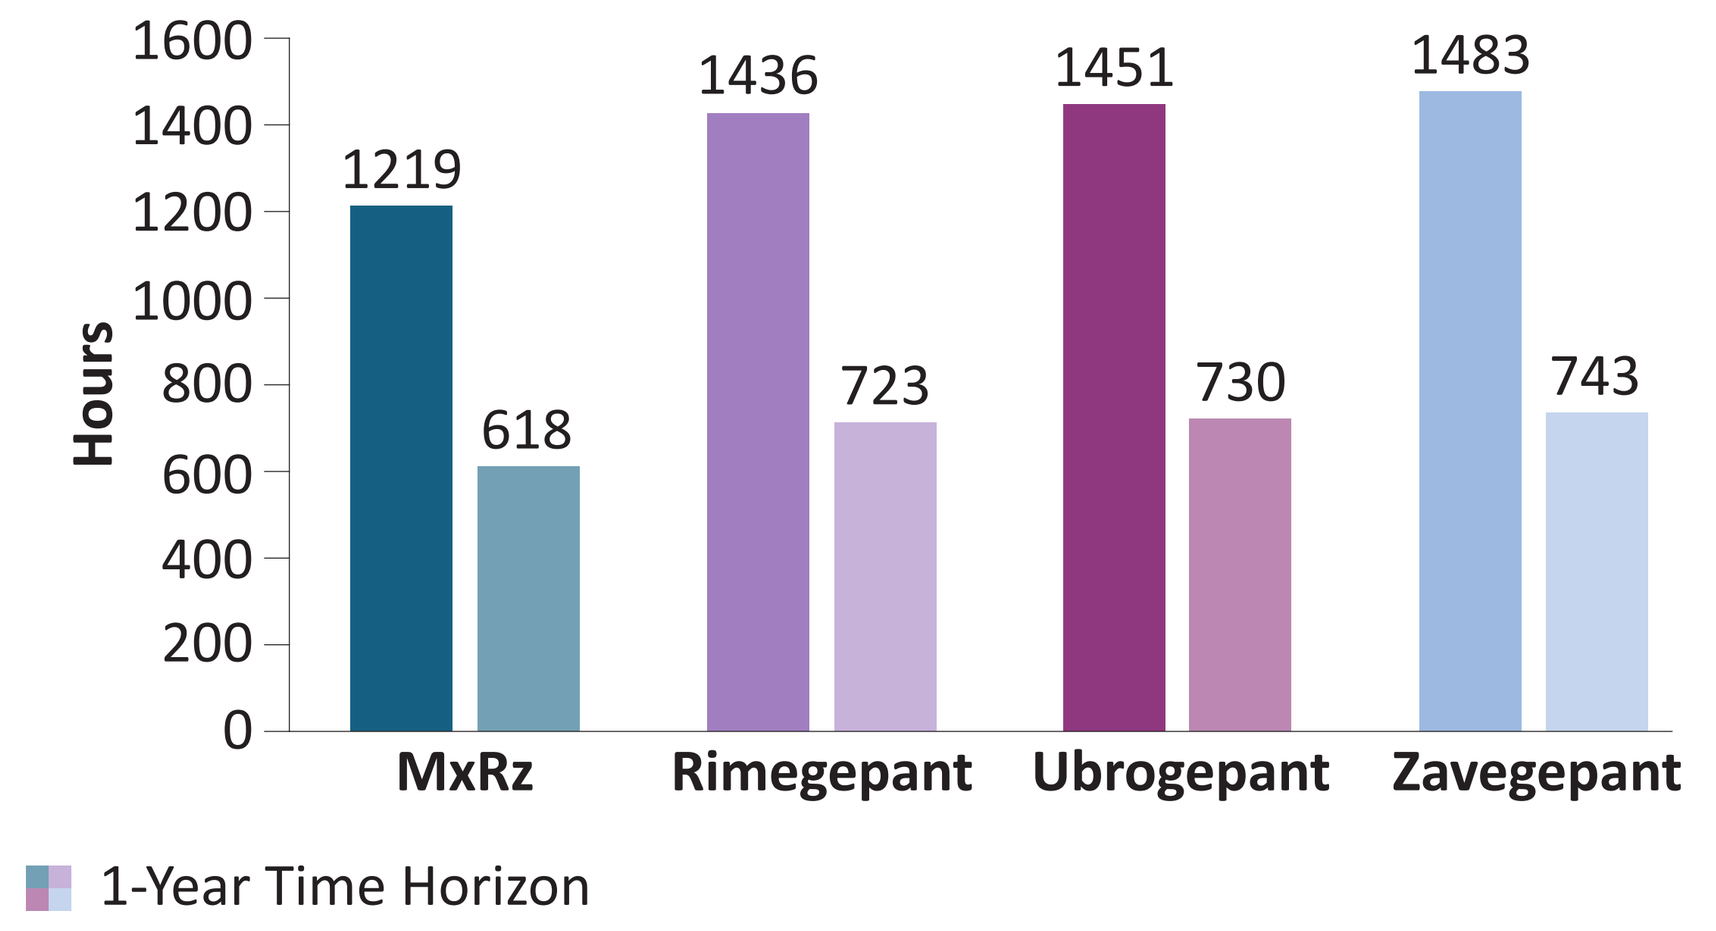
Hours
1600
1400
1200
1000
800
600
400
200
0
1219 618
1436 723
1451 730
1483 743
MxRz
Rimegepant
Ubrogepant
Zavegepant
2-Year Time Horizon
1-Year Time Horizon

- MxRz generated slightly higher QALYs (1.860) and fewer hours of pain (1219) than rimegepant (1.855 QALYs/1436 hours), ubrogepant (1.855 QALYs/1451 hours), or zavegepant (1.853 QALYs/1483 hours), making it the dominant intervention from a US payer perspective
- Similar trends were observed in 1-year analyses
- Sensitivity analyses varying base-case values (OWSA) or parameters (PSA) confirmed the model's robustness
- In the PSA with 1000 iterations, MxRz remained dominant when compared with each gepant, suggesting better health outcomes and lower total costs

Note: Estimated total QALYs and hours of pain were derived from the NMA

View previous NMA poster

Cost Per Pain Freedom Responder and Rescue Medication Avoided

Cost per Responder

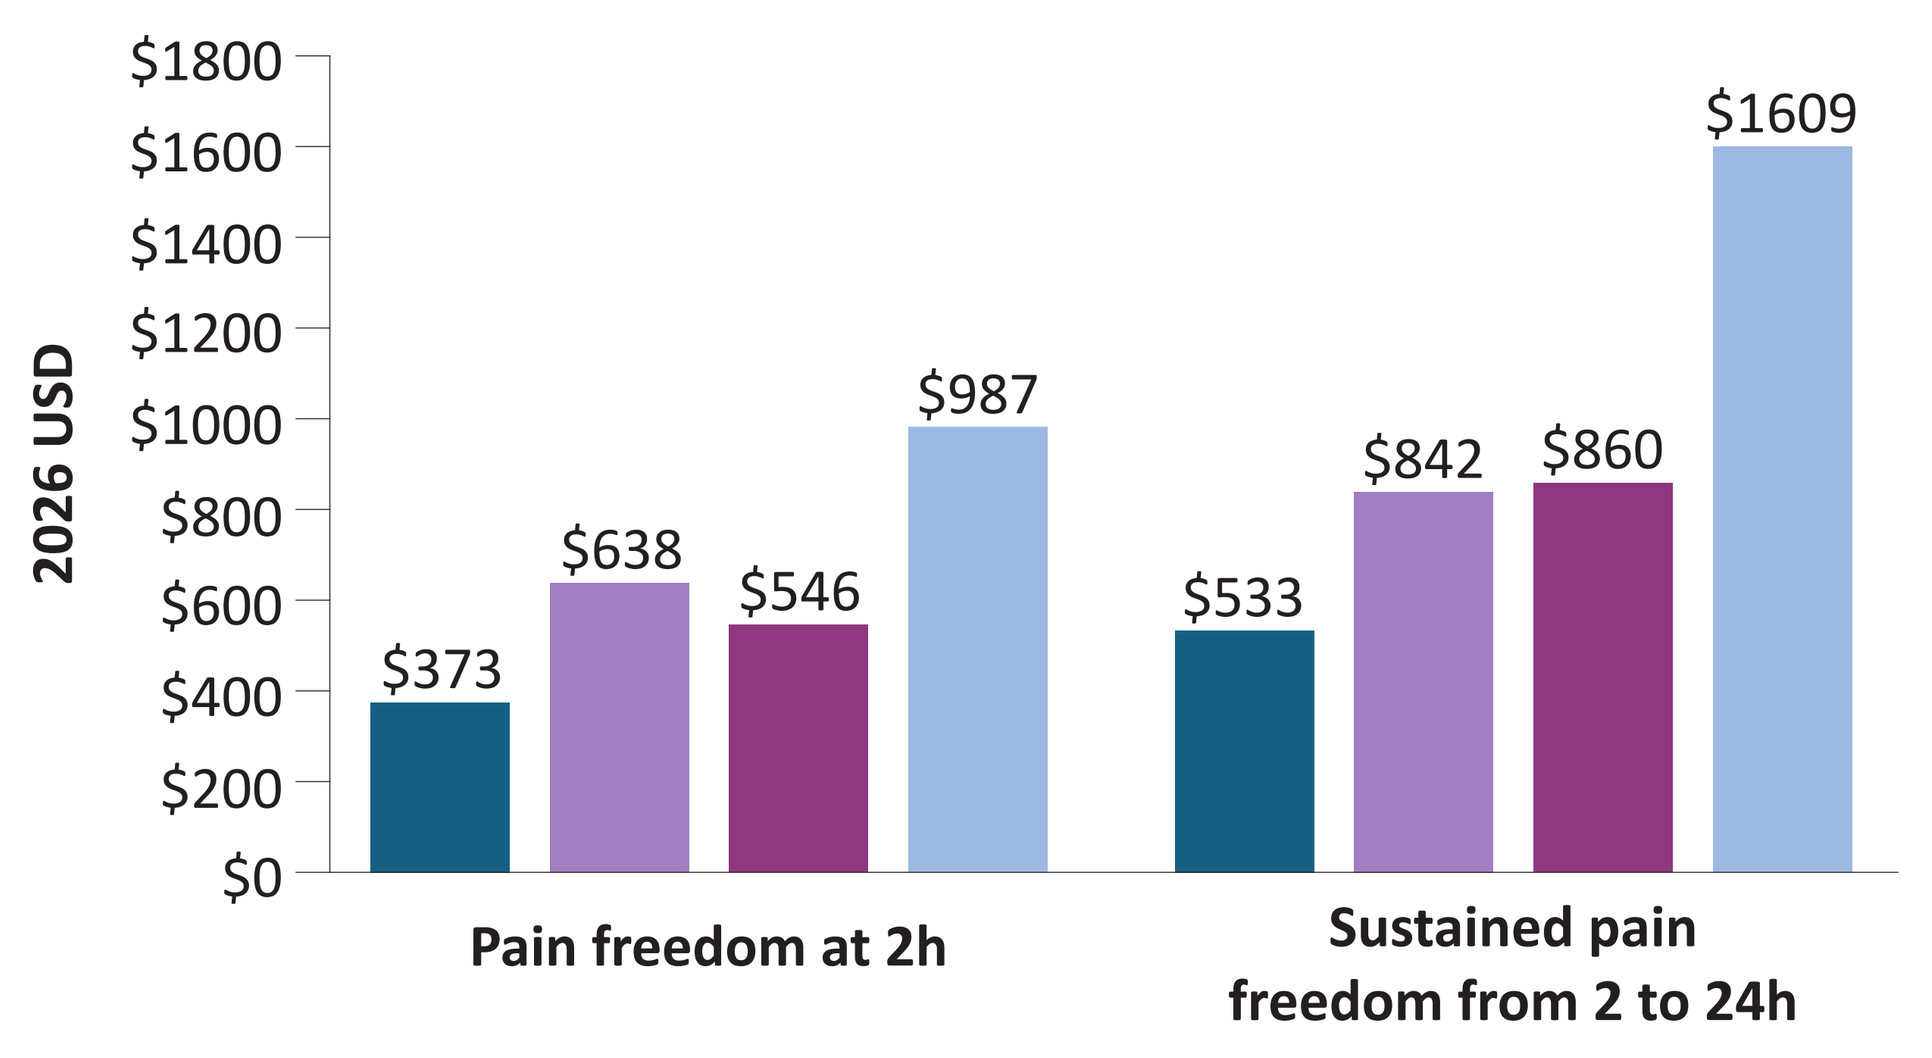
2026 USD
$1800
$1600
$1400
$1200
$1000
$800
$600
$400
$200
$0
$373
$638
$546
$987
$533
$842
$860
$1609
Pain freedom at 2h
Sustained pain freedom from 2 to 24h

Cost per Rescue Medication Avoided

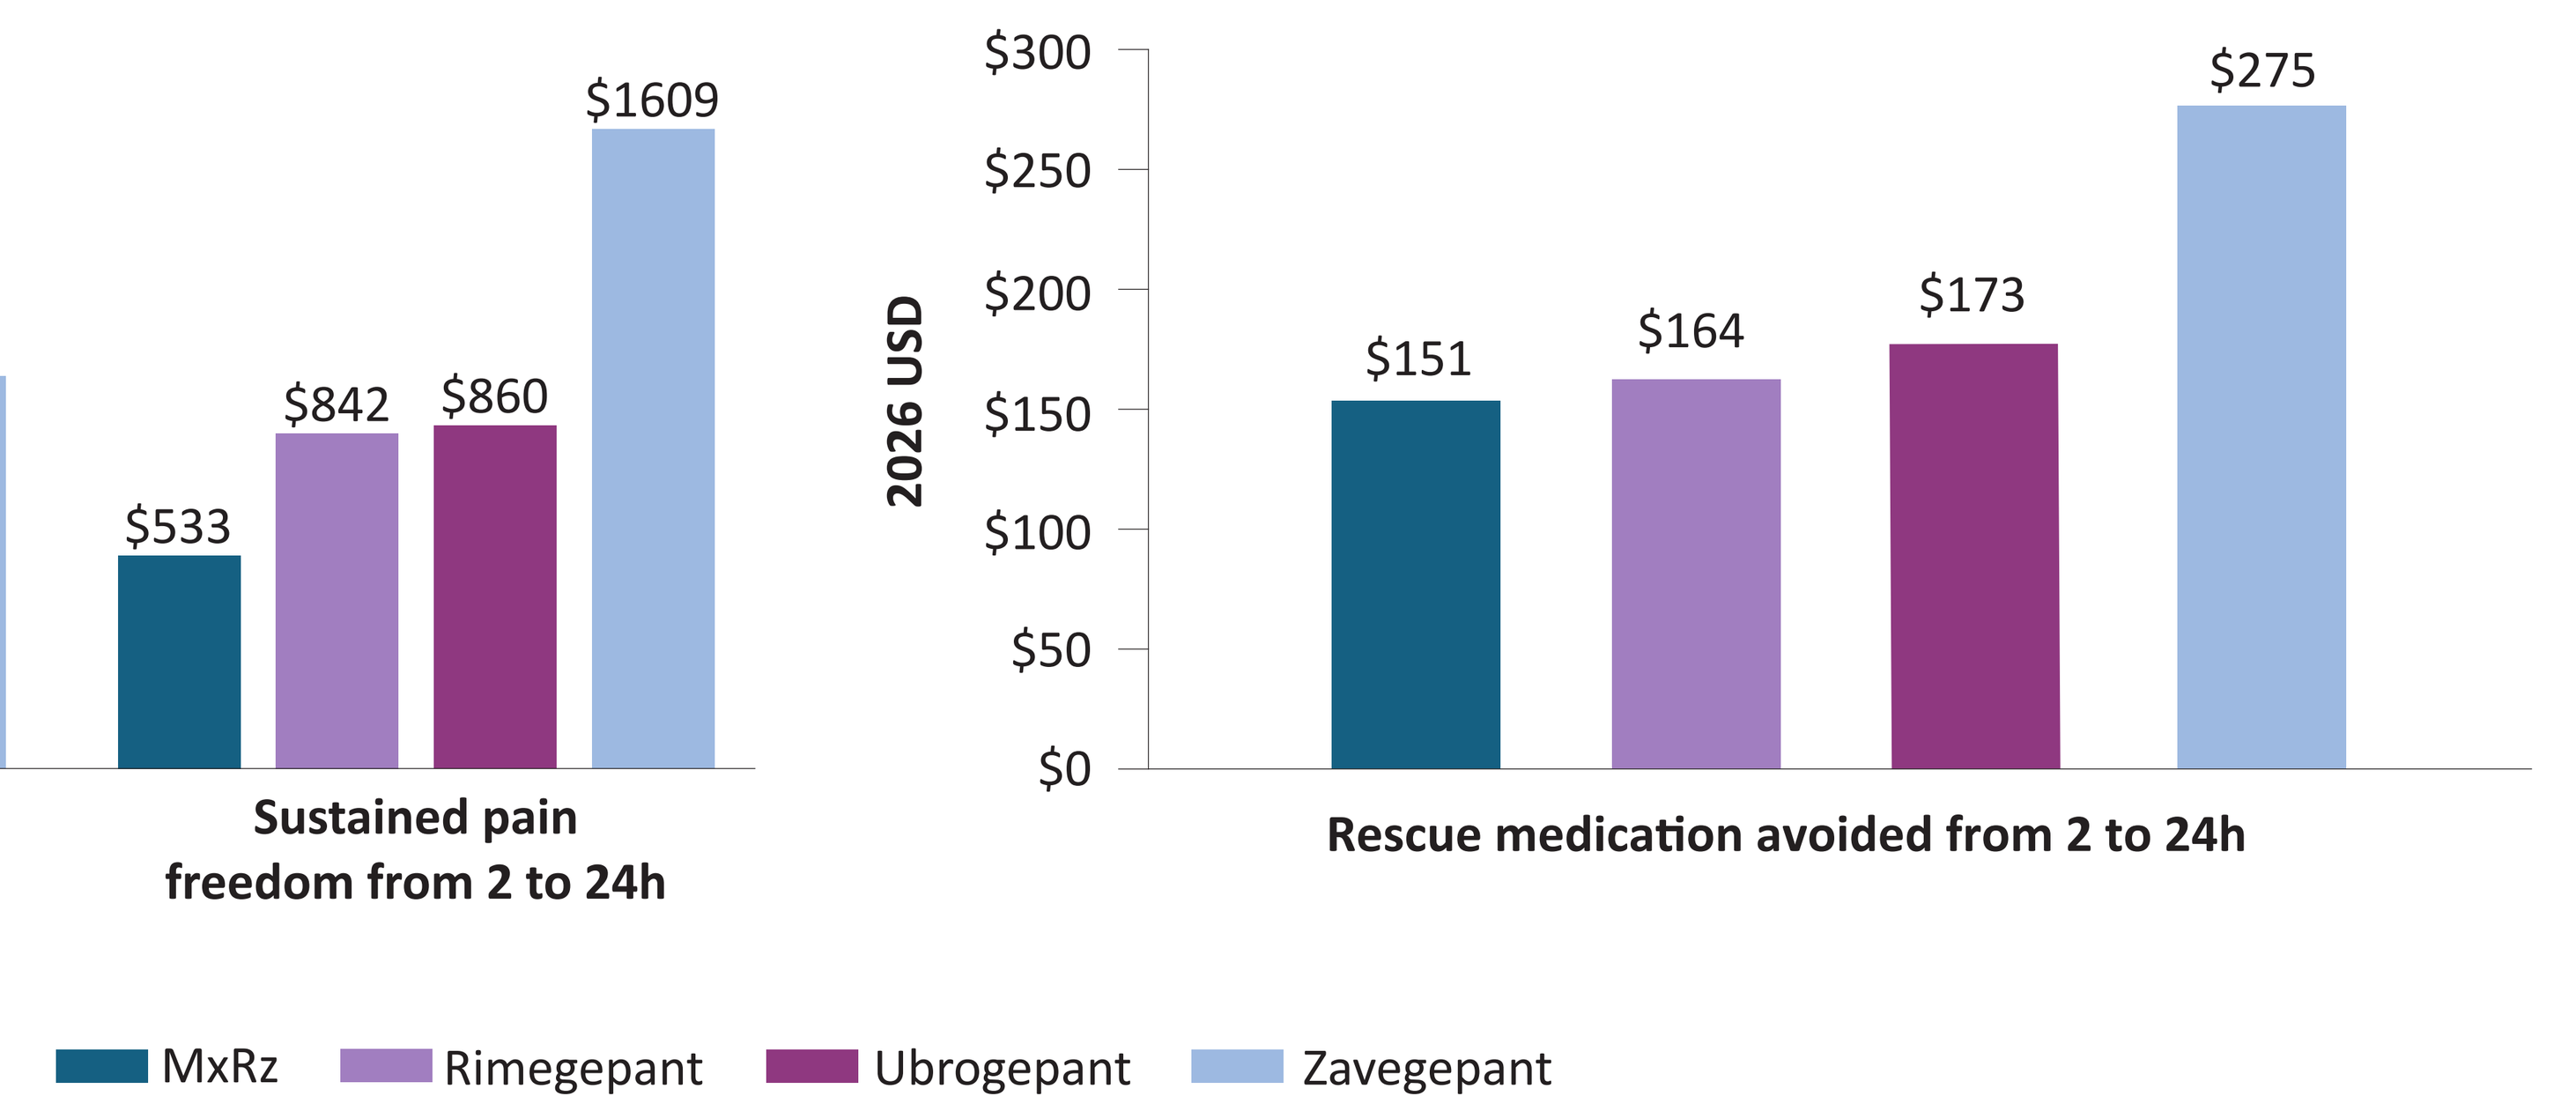
2026 USD
$300
$250
$200
$150
$100
$50
$0
$151
$164
$173
$275
Rescue medication avoided from 2 to 24h
MxRz
Rimegepant
Ubrogepant
Zavegepant

- MxRz demonstrated the lowest costs per responder for pain freedom at 2 hours, cost per responder for sustained pain freedom from 2 to 24 hours, and per rescue medication avoided from 2 to 24 hours—followed by ubrogepant, rimegepant, and zavegepant

Limitations

- Drug unit costs were based on 2026 wholesale acquisition costs, which may not reflect actual costs
- Treatment efficacy was derived from NMA results and may be influenced by study heterogeneity, differences in patient population, and variations in study design
- The model extrapolated 48-hour pain trajectories based on 2-hour pain relief and pain freedom outcomes, which may not fully capture longer-term clinical response patterns

Conclusions

- In an environment of rising costs associated with treatment of migraine, MxRz is assessed to be cost-effective when compared with gepants (rimegepant, ubrogepant, and zavegepant) from a US payer perspective
- Based on the NMA model-derived outcomes, treatment with MxRz was found to be associated with lower total costs, higher QALYs, and lower costs per responder and per rescue medication avoided when compared with each gepant

References
1. Tepper SJ. Neurol Clin. 2019;37:727-42.
2. Morton BA, et al. https://headachemigraine.org/wp-content/uploads/2025/02/CHAMP-Survey-Brief-2.pdf.
3. Smelt AF, et al. PLoS One. 2014;9(6):e98933.
4. Irimia P. J Headache Pain. 2022;23(1):78.
5. Lipton RB, et al. Neurology. 2015;84:688-95.
6. Lipton RB, et al. Headache. 2016;56:1635-48.
7. Marcus SC, et al. Cephalalgia. 2020;40:639-49.
8. Shewale AR, et al. Cephalalgia. 2024;44(2):3331024241235139.
9. Younis S, et al. Handb Clin Neurol. 2024;199:51-66.
10. Jones A, et al. Neurology. 2022;98(Suppl 18):3564.
11. Symbravo [package insert]. Axsome Therapeutics. January 2025. https://www.axsome.com/wp-content/uploads/2025/02/SYM-USPI-001.001-20250224.pdf.
12. Lipton RB, et al. J Pain Res. 2024;17:2431-41.
13. Lipton RB, et al. N Engl J Med. 2019;381(2):142-9.
14. Croop R, et al. Lancet. 2019;394(10200):737-45.
15. Dodick DW, et al. N Engl J Med. 2019;381(23):2230-41.
16. Lipton RB, et al. JAMA. 2019;322(9):1887-98.
17. Hutchinson S, et al. Neurol Ther. 2021;10:235-49.
18. Croop R, et al. Headache. 2022;62(9):1153-63.
19. Lipton RB, et al. Lancet Neurol. 2023;22(3):209-17.
20. Nahas J, et al. Comparative efficacy of AXS-07 versus gepants for acute treatment of migraine: a network meta-analysis. Poster presented at the AMCP Annual Meeting; 31 March-3 April 2025. Houston, TX.
21. Nguyen JL, et al. J Headache Pain. 2022;23(1):111.
22. Atlas S, et al. 2020. https://icer.org/wp-content/uploads/2020/11/ICER_Acute-Migraine_Final-Evidence-Report_092221.pdf.
23. Jones A, et al. Treatment of migraine pain and associated symptoms with AXS-07: Results from MOVEMENT, a long-term efficacy and safety study. Poster presented at the American Headache Society Virtual Annual Scientific Meeting; 3-6 June 2021.
24. Mullin K, et al. Cephalalgia. 2024;44(8):3331024241259456.
25. Silberstein SD, et al. Headache. 2018;58(10):1579-92.
26. Bureau of Economic Analysis, US Department of Commerce. 2024. https://www.bea.gov/data/personal-consumption-expenditures-price-index.
27. Merative Micromedex. 2025. https://www.merative.com/clinical-decision-support/micromedex-redbook.
28. Rimegepant PI. 2020. https://labeling.pfizer.com/ShowLabeling.aspx?id=19036.
29. Ubrogepant PI. 2019. https://www.accessdata.fda.gov/drugsatfda_docs/label/2019/211765s000lbl.pdf.
30. Zavegepant PI. 2023. https://www.accessdata.fda.gov/drugsatfda_docs/label/2023/216386s000lbl.pdf.

Disclosures
CG and AE are full-time employees of RTI Health Solutions, an independent nonprofit research organization, which was retained by Axsome Therapeutics to conduct the research that is the subject of this poster. Their compensation is unconnected to the studies on which they work. YZ, TG, and RD are employees of Axsome Therapeutics and may hold shares and/or stock options in the company. JA has consulted for AbbVie, Aeon, Axsome, Amneal, Dr. Reddy, Eli Lilly, Lundbeck, Linpharma, Ipsen, Merz, Pfizer, Gore, Satsuma, and Vectura Fertin; received grants from Mi-Helper (PI) and Parema, Ipsen, Lundbeck, Pfizer, and Merz (institution); and serves as a medical editor on the editorial board/steering committee for SELF magazine. This research was funded by Axsome Therapeutics as part of a research contract with RTI Health Solutions. Axsome Therapeutics provided funding for publication support in the form of writing, styling, and submission. No honoraria or payments were made for authorship.

Contact Information
Yang Zhao
Axsome Therapeutics
One World Trade Center, 22nd floor,
New York, NY, 10007
1.917.692.0120
yzhao@axsome.com

QR Code
Scan the QR code at the top of this poster or access **https://www.axsomecongresshub.com/AMCPNexus2026.shtml** to view or download a PDF of this poster or access additional information.