American Society of Consultant Pharmacists
October 29 – November 1, 2026
National Harbor, MD, USA

Agitation in Alzheimer's Disease: Association with Increased Clinical Burden, Healthcare Utilization, and Mortality Among Patients in Long-Term Care

Manju T Beier,[1] Yang Zhao,[2*] Zhishui Zheng,[3] Jennifer Friderici,[3] Courtney Zeni,[2] Michael Tocco,[2] Ronnie DePue,[2] Malcolm Fraser[4]

[1]Geriatric Consultant Resources LLC, Ann Arbor, MI, USA and University of Michigan, Ann Arbor, MI, USA; [2]Axsome Therapeutics, Inc., New York, NY, USA; [3]KMK Consulting, Inc., Morris Plains, NJ, USA; [4]Red Rock Pharmacy, Salt Lake City, UT, USA

Key Objective

To characterize the real-world clinical burden, healthcare resource utilization, medication use, mortality, and cognitive function among long-term care residents with Alzheimer's disease/dementia and agitation compared with matched residents without agitation

Introduction

- Agitation associated with dementia due to Alzheimer's disease (AD) is a common, debilitating neuropsychiatric symptom that drives substantial patient and caregiver burden, especially in long-term care (LTC) settings[1-3]
- Pharmacologic treatment commonly relies on off-label antidepressants and antipsychotics, with variable effectiveness and associated safety concerns[4,5]
- At the time of this analysis, the atypical antipsychotic brexpiprazole was the only US Food and Drug Administration-approved treatment for agitation in AD[6]
- Although a substantial proportion of patients with agitation associated with AD reside in LTC as severity and care needs increase, real-world data on the impact of agitation in this setting remain limited[7]

Methods

Study Design

- **Data source and study period** — PointClickCare electronic health record dataset of LTC residents; data collected from June 2020 to May 2025
- **Agitation cohort** — Adults with ≥ 1 diagnosis of dementia or AD and ≥ 1 diagnosis or indicator of agitation; index date = first recorded agitation event between June 1, 2020 and May 31, 2024
- **Matched non-agitation cohort** — Adults with ≥ 1 diagnosis of dementia or AD and no evidence of agitation during the 12-month pre-index and post-index periods. Index dates were randomly assigned and patients were matched 1:1 to the agitation cohort on age, sex, race, and US geographic region
- **Study eligibility** — Continuous record availability during the 12-month pre- and post-index periods. Residents with missing or unknown sex, race, or US geographic region or other data quality issues were excluded

Outcomes

- **Clinical burden** — Comorbidities, falls, and harm events to self or others (12-month post-index period)
- **Healthcare resource utilization (HCRU) and medication use** — Emergency room (ER) visits, hospital admissions, and medication use (12-month post-index period)
- **Mortality** — From the end of the 12-month post-index period through the last available record
- **Cognitive function** — Brief Interview for Mental Status (BIMS) scores assessed from index through the last available record

Statistical Analysis

- **Descriptive statistics** — Means (SD) are reported for continuous variables and frequencies (%) for categorical variables
- **Post-index outcomes**—Models accounted for the matched cohort design and adjusted for index year and selected baseline comorbidities; cognitive analyses also adjusted for baseline BIMS score. Binary probability outcomes used conditional logistic regression; count outcomes used generalized estimating equations with a Poisson distribution. Predicted values were estimated from fitted models
- **Effect estimates** —Odds ratios (ORs) or incidence rate ratios (IRRs), with 95% confidence intervals (CIs); $P < 0.05$ indicated statistical significance
- **Mortality** — Kaplan-Meier analyses with adjusted hazard ratios (HRs) estimated using Cox proportional hazards models controlling for index year and selected baseline comorbidities

References

1. Grossberg G, et al. *J Med Econ*. 2024;27(1):99–108. 2. Van der Mussele S, et al. *Aging Ment Health*. 2015;19(3):247–257. 3. Carrarini C, et al. *Front Neurol*. 2021;12:644317. 4. McDermott CL, Gruenewald DA. *Curr Geriatr Rep*. 2019;8(1):1–11. 5. Teixeira AL, et al. *World J Psychiatry*. 2025;15(11):109581. 6. REXULTI [Full Prescribing Information]. Rockville, MD: Otsuka America Pharmaceutical, Inc. 7. Arrighi HM, et al. *Alzheimer Dis Assoc Disord*. 2010;24(1):90–95.

Acknowledgments

This study was funded by Axsome Therapeutics, Inc. Medical writing and editorial support was provided by Asclepius Analytics and was funded by Axsome Therapeutics.

QR Code

Scan the QR code at the top of this poster or access **https://www.axsomecongresshub.com/ASCPConsultantPharmacists2026.shtml** to view or download a PDF of this poster or access additional information and other Axsome Therapeutics presentations

Disclosures

M Fraser serves as Medical Director for 9 Nursing Homes; consultant and/or speaker for Paradigm Consulting, AFPA (Alliance for Patient Access), Red Rock Pharmacy, Acadia, Alpha Cognition, Axsome Therapeutics, and Shionogi; IAB Co-Chair and Medical Affairs Advisor for Behavior Health Solutions; member of the State of Colorado Chapter 5 Rewrite Committee and the PaltMed CMO Special Interest Group; and Medical Director/Editor of the Red Rock Report. Prior relationships include Neurocrine, Urovant, Otsuka, and Lilly.
Y Zhao, C Zeni, M Tocco, and R DePue are employees of Axsome Therapeutics, Inc.
Z Zheng and J Friderici are employees of KMK Consulting, Inc.
MT Beier has served on speaker bureaus and as a consultant for Axsome Therapeutics, Inc.

Results

Baseline Patient Characteristics

	Agitation cohort N = 126,667	Non-agitation cohort N = 126,667
Age, years, mean (SD)	80.6 (7.4)	80.6 (7.4)
Female, %	70.2%	70.2%
Race, %		
Black or African American	12.5%	12.5%
White	80.7%	80.7%
Other	6.8%	6.8%
US geographic region, %		
Northeast	21.9%	21.9%
Midwest	29.8%	29.8%
South	35.6%	35.6%
West	12.8%	12.8%
Comorbidities, %		
Anxiety	24.8%	17.2%
Depression (not bipolar)	38.2%	30.5%
Hypertension	53.4%	47.4%
Falls		
Mean number (SD)	0.84 (2.33)	0.58 (1.83)
Patients, %	26.5%	21.3%

SD, standard deviation; US, United States.

- Residents with agitation had a greater pre-index clinical burden, including higher rates of anxiety, depression, hypertension, and falls. Harm events, ER visits, and hospital admissions were similar between cohorts (data not shown)

Comorbidities

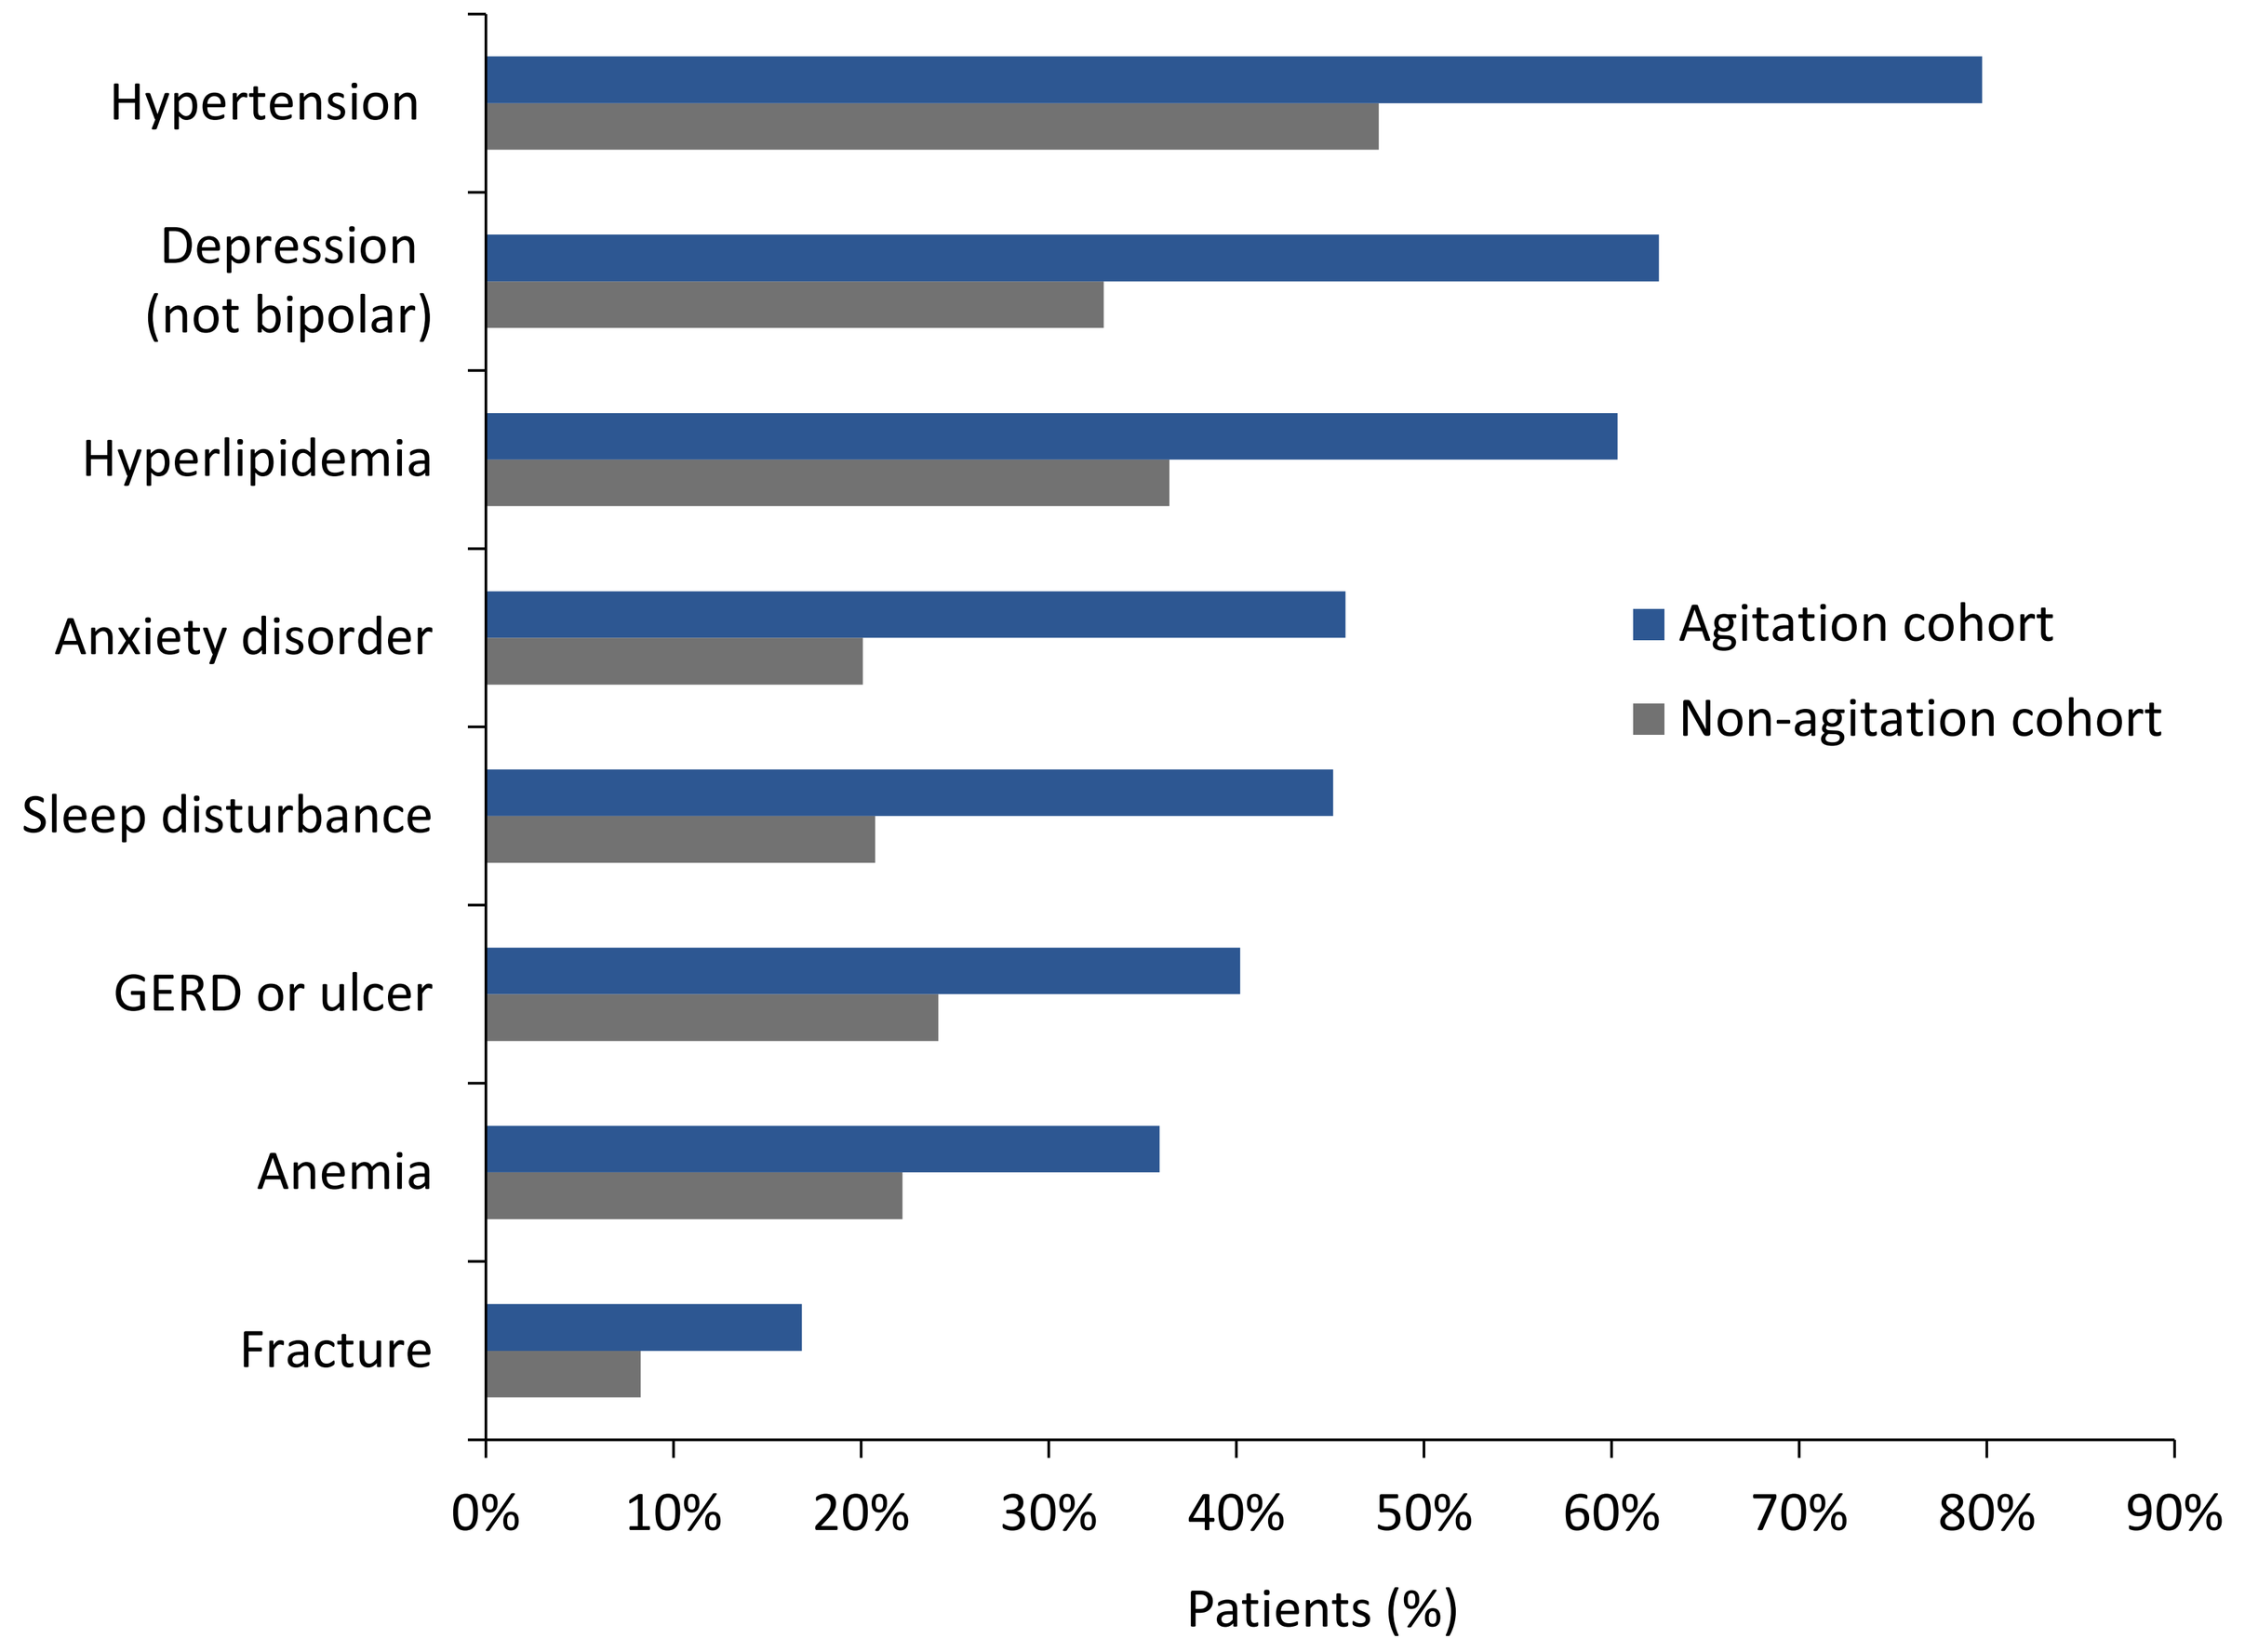
Hypertension
Depression (not bipolar)
Hyperlipidemia
Anxiety disorder
Sleep disturbance
GERD or ulcer
Anemia
Fracture
Agitation cohort
Non-agitation cohort
0%
10%
20%
30%
40%
50%
60%
70%
80%
90%
Patients (%)

Comorbidities examined across classes of cardiovascular, cerebrovascular, hematologic, infectious diseases, metabolic, neurologic, psychiatric, renal, respiratory, sensory, musculoskeletal, sleep disorders, gastrointestinal, and genitourinary disorders. Selected comorbidities of particular relevance to patients with Alzheimer's disease are presented.
GERD, gastroesophageal reflux disease.

- Comorbidities were significantly more prevalent in the Agitation cohort during the 12-month post-index period, with 1.45- to 3.79-fold higher adjusted prevalence than in the Non-agitation cohort across all evaluated comorbidity classes

Falls, Harm Events, and HCRU

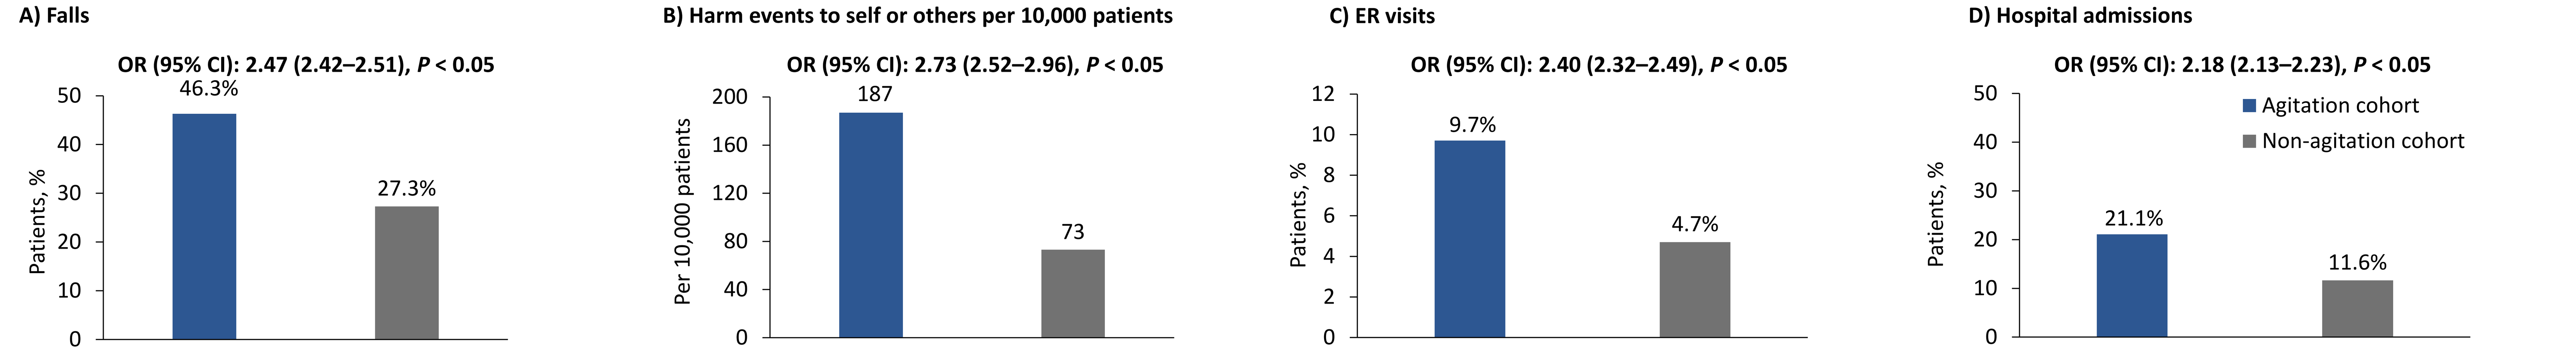
A) Falls
OR (95% CI): 2.47 (2.42–2.51), P < 0.05
Patients, %
46.3%
27.3%
B) Harm events to self or others per 10,000 patients
OR (95% CI): 2.73 (2.52–2.96), P < 0.05
Per 10,000 patients
187
73
C) ER visits
OR (95% CI): 2.40 (2.32–2.49), P < 0.05
Patients, %
9.7%
4.7%
D) Hospital admissions
OR (95% CI): 2.18 (2.13–2.23), P < 0.05
Patients, %
21.1%
11.6%
Agitation cohort
Non-agitation cohort

CI, confidence interval; ER, emergency room; OR, odds ratio.

- Agitation was associated with approximately 2- to 3-fold higher odds of falls, harm events, ER visits, and hospital admissions among LTC residents with AD/dementia. Agitation was also associated with a significantly higher mean number of falls, harm events, ER visits, and hospital admissions (IRRs 1.97–2.63; all $P < 0.05$; data not shown)

Mortality

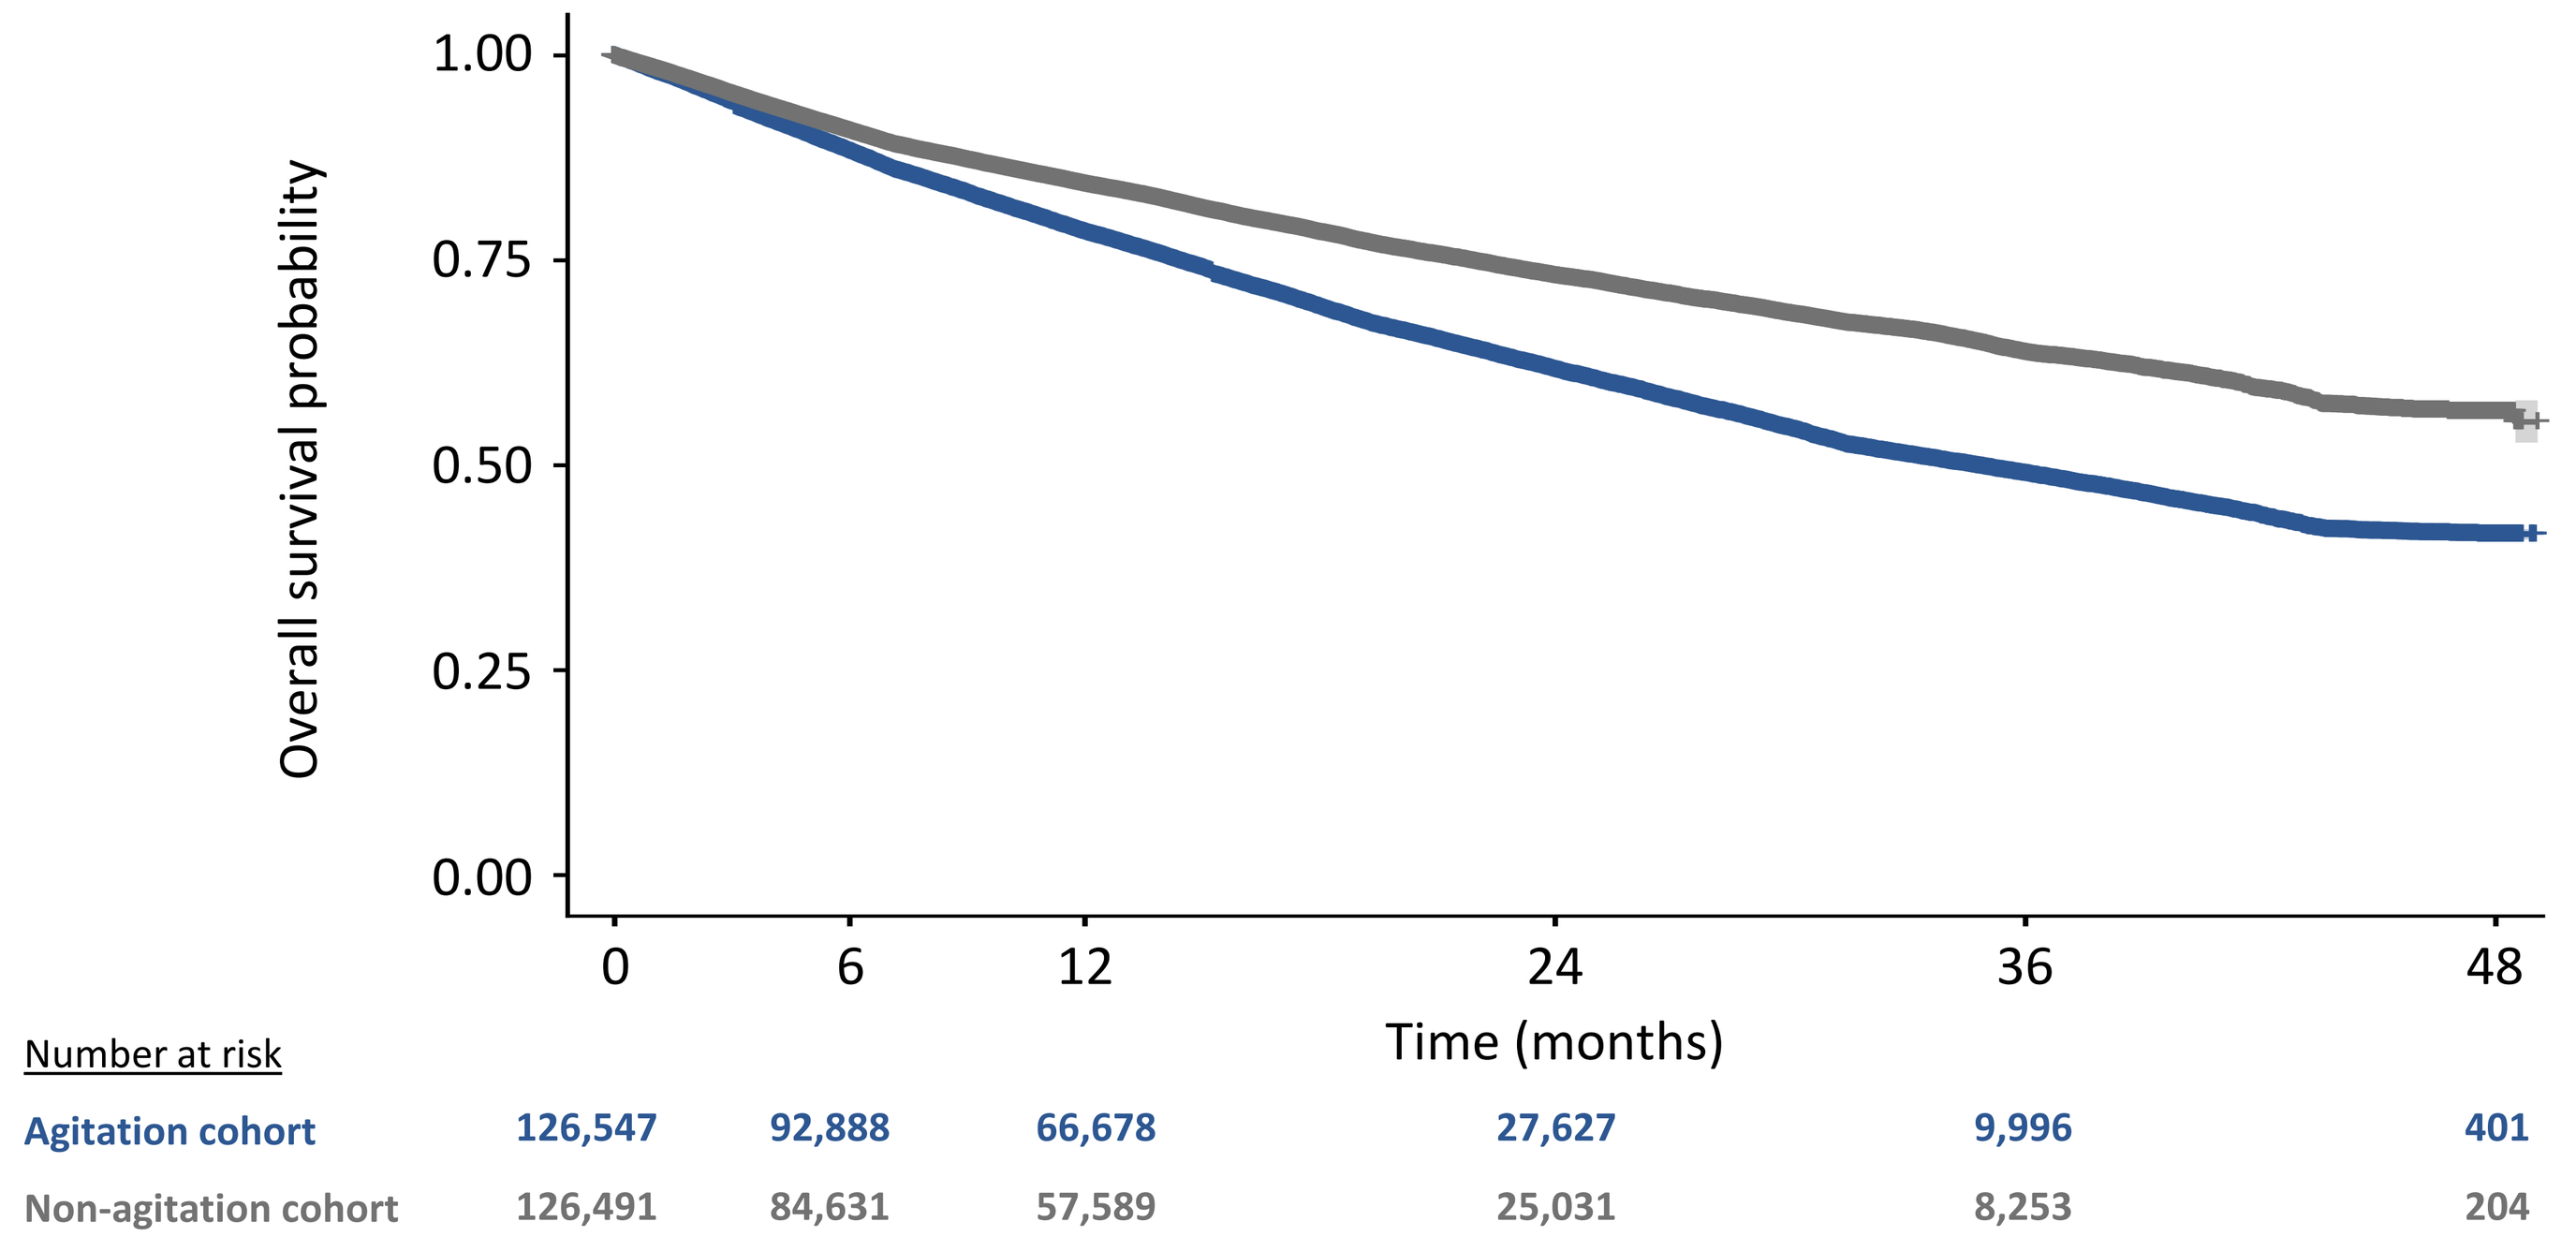
Overall survival probability
1.00
0.75
0.50
0.25
0.00
0
6
12
24
36
48
Time (months)
Number at risk
Agitation cohort 126,547 92,888 66,678 27,627 9,996 401
Non-agitation cohort 126,491 84,631 57,589 25,031 8,253 204

Timepoint 0 = end of the 12-month post-index period.

- Agitation was associated with a significantly increased risk of mortality, with an approximately 40% higher adjusted risk of death that remained elevated throughout 48 months of follow-up (HR 1.39, 95% CI 1.36–1.42)

Cognitive Function

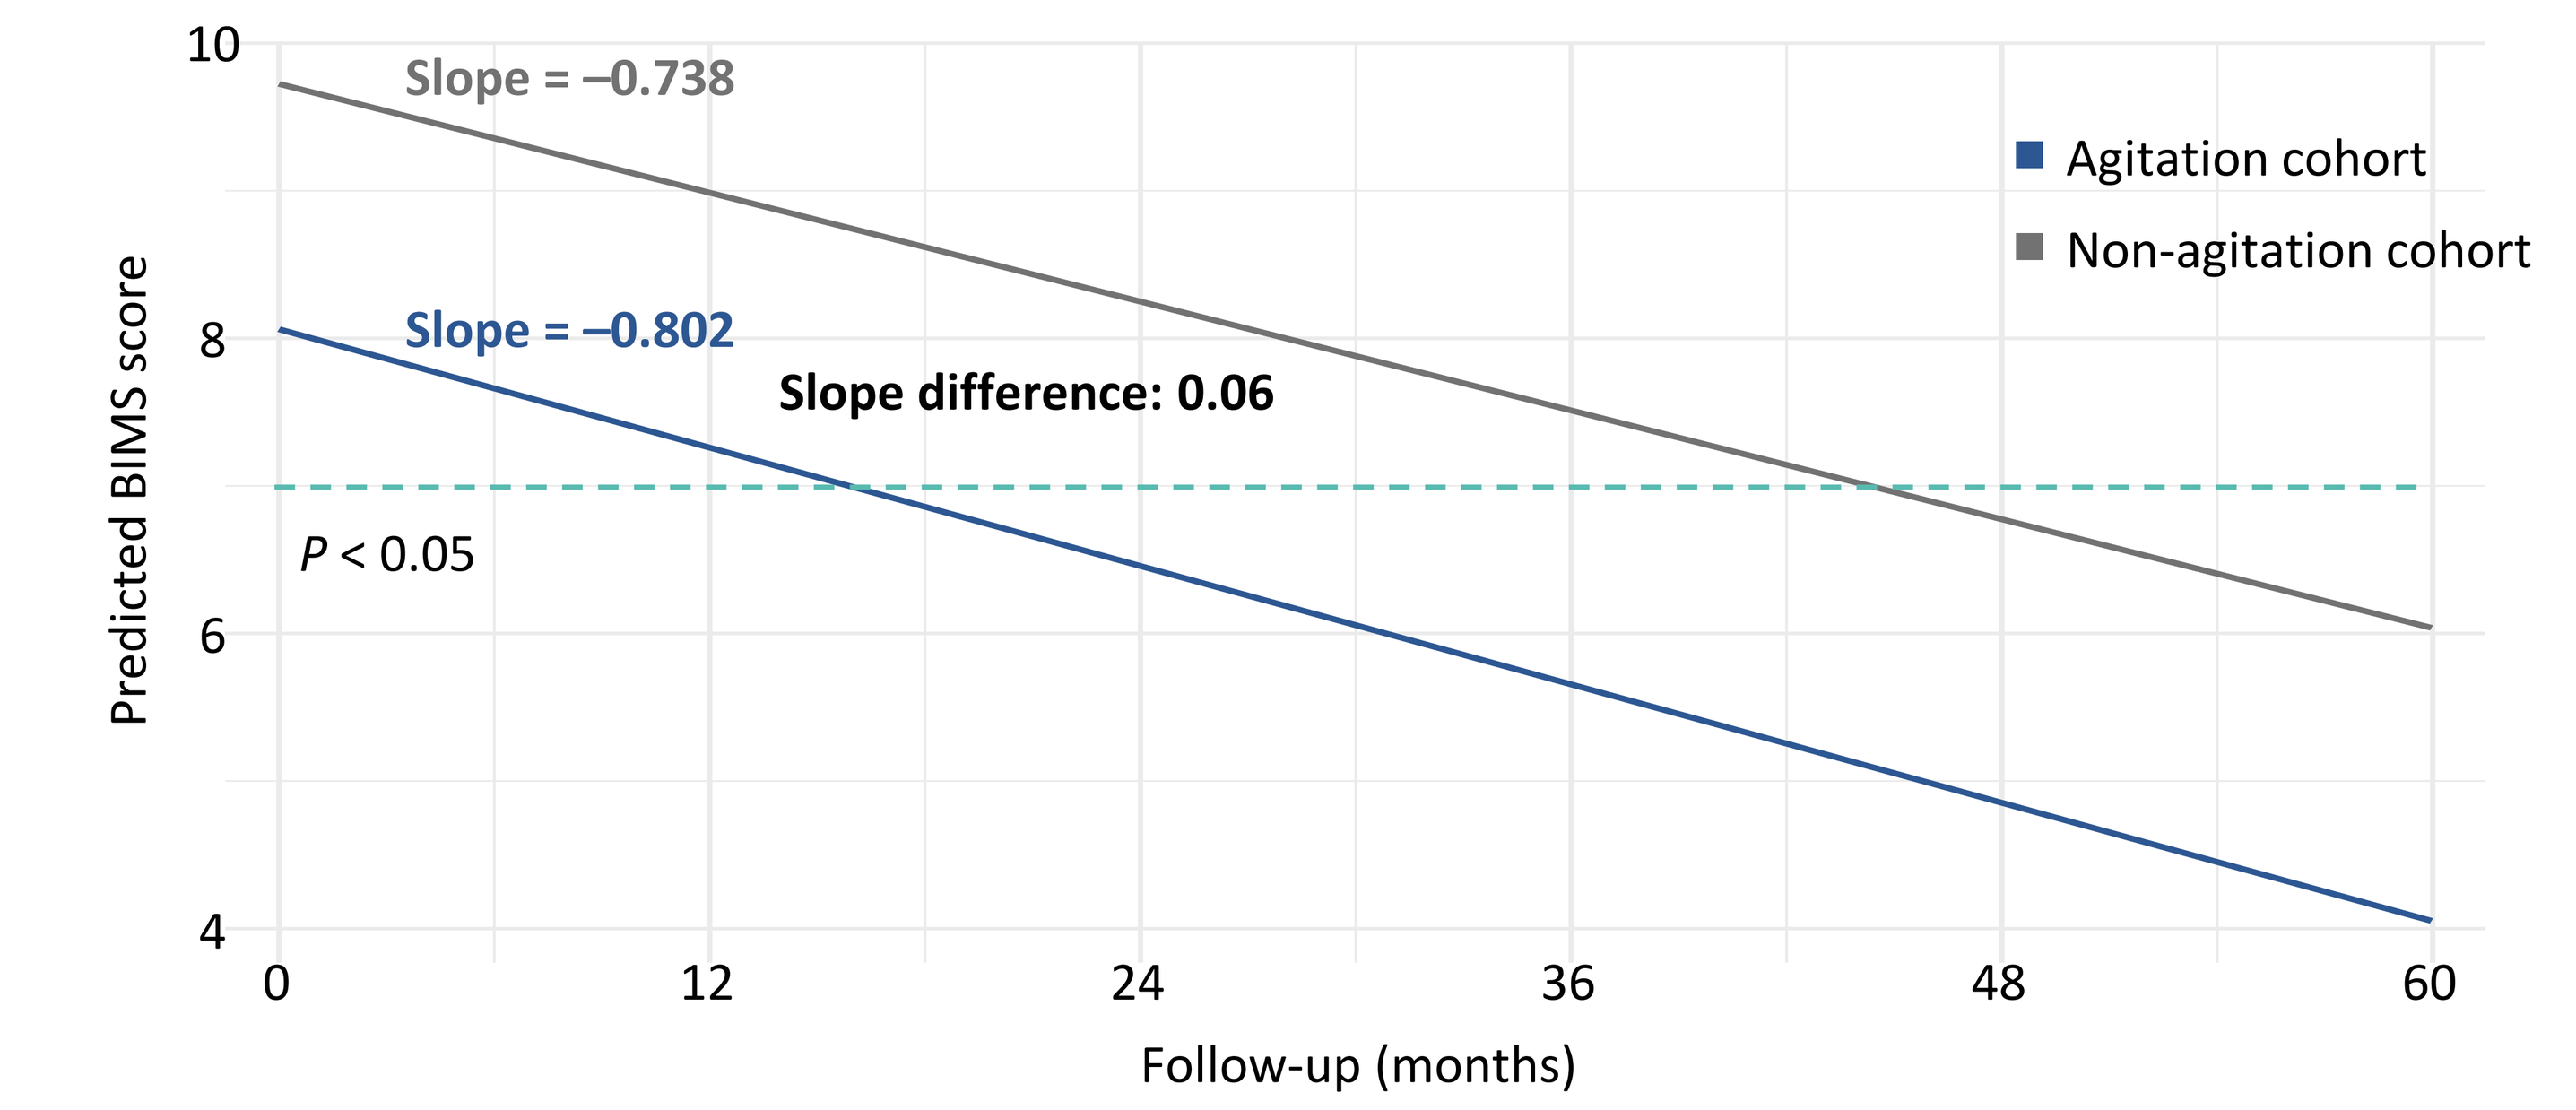
Slope = –0.738
Slope = –0.802
Slope difference: 0.06
P < 0.05
Agitation cohort
Non-agitation cohort
Predicted BIMS score
Follow-up (months)

Dashed line indicates BIMS = 7; BIMS 0–7, severe impairment; 8–12, moderate impairment.
BIMS, Brief Interview for Mental Status.

- Agitation was associated with faster cognitive decline, with adjusted BIMS scores declining approximately 9% more rapidly and remaining consistently lower throughout follow-up

Medication Use

	Agitation cohort N = 126,667	Non-agitation cohort N = 126,667	Estimate (95% CI)
Antidepressants			
Predicted probability	0.61	0.41	OR 2.34 (2.30–2.39)
Mean number of distinct agents	0.88	0.55	IRR 1.58 (1.56–1.59)
Antipsychotics			
Predicted probability	0.31	0.11	OR 3.91 (3.82–4.01)
Mean number of distinct agents	0.36	0.12	IRR 3.03 (2.98–3.09)
Benzodiazepines, overall			
Predicted probability	0.18	0.08	OR 2.53 (2.46–2.60)
Mean number of uses	27.74	10.68	IRR 2.34 (2.22–2.46)
Benzodiazepines, PRN			
Predicted probability	0.10	0.04	OR 2.68 (2.59–2.77)
Mean number of uses	2.24	0.87	IRR 2.74 (2.72–2.76)

CI, confidence interval; IRR, incidence rate ratio; OR, odds ratio; PRN, *pro re nata* (use as needed).

- Medication use was significantly greater in the Agitation cohort during the 12-month post-index period, including antidepressants, antipsychotics, and benzodiazepines (overall and PRN)
- Patients with agitation were more likely to receive these medications and had greater medication intensity, as reflected by higher numbers of distinct agents and benzodiazepine uses

Limitations

- Despite 1:1 matching and covariate adjustment, residual confounding may persist
- Agitation was identified from diagnoses and clinical indicators in medical records; severity could not be assessed, and misclassification was also possible
- The dataset lacked information on treatment indication, dosing, and adherence; therefore, treatment selection could not be attributed to specific diagnoses or symptoms
- Survival was assessed after the 12-month post-index period, with potential introduction of survivor bias
- The findings reflect residents of US LTC facilities using the PointClickCare system and may not extend to other settings

Conclusions

- Among matched US LTC residents with AD/dementia, agitation was associated with a substantially greater clinical burden across all outcomes assessed, including:
 - More comorbidities
 - More falls and harm events
 - More ER visits and hospital admissions
 - Greater use of antipsychotics, antidepressants, and benzodiazepines
 - Higher mortality risk and accelerated cognitive decline
- These findings suggest that agitation may confer a substantial clinical and safety risk, highlighting the need for effective, well-tolerated treatments and proactive care strategies that enable earlier identification and management of agitation in the LTC setting